A LONG TERM ANALYSIS OF THE CLINICAL AND COST EFFECTIVENESS OF GLATIRAMER ACETATE FROM THE UK MULTIPLE SCLEROSIS RISK SHARING SCHEME

PND98

Montu Sumra[1], John R Fullarton[2], Ewan Walters[1]

[1] Medical Department, Teva UK Limited, Castleford, UK

[2] Strategen Limited, Basingstoke, UK

ISPOR 18th Annual European Congress· 7th-11th November 2015 · Milan, Italy

INTRODUCTION

- The UK Multiple Sclerosis Risk Sharing Scheme (RSS) was established in 2002 to provide UK patients access to the disease-modifying therapies (DMTs) glatiramer acetate (GA) and the beta interferons, and, in the form of a 10-year observational study, monitored a cohort of them to evaluate the long term effectiveness of these therapies[1]
- A continuous Markov model is used to assess whether disability progression on each of the DMTs is consistent with a cost effectiveness target of £36,000 per quality-adjusted life-year (QALY) projected over 20 years (as stipulated at the outset of the Scheme)
- Projections are made with the model using natural history data from the British Columbia Multiple Sclerosis database (BCMS) and hazard ratios (HRs) for attenuated disability progression, as measured by Expanded Disability Status Scale (EDSS) score, derived from the individual randomised controlled trials (RCTs) of each drug
- Actual disability progression was monitored in a cohort of 5,600 patients, with the intention of making price adjustments to maintain cost effectiveness should their progress differ substantially from the projections
- 6-year data from the RSS has recently been published and show that the DMTs, when analysed together, were clinically and cost effective when modelled over the 20-year time horizon (Figure 1)[2]

Figure 1: Progression and utility ratios for 6-year RSS data in aggregate[2]

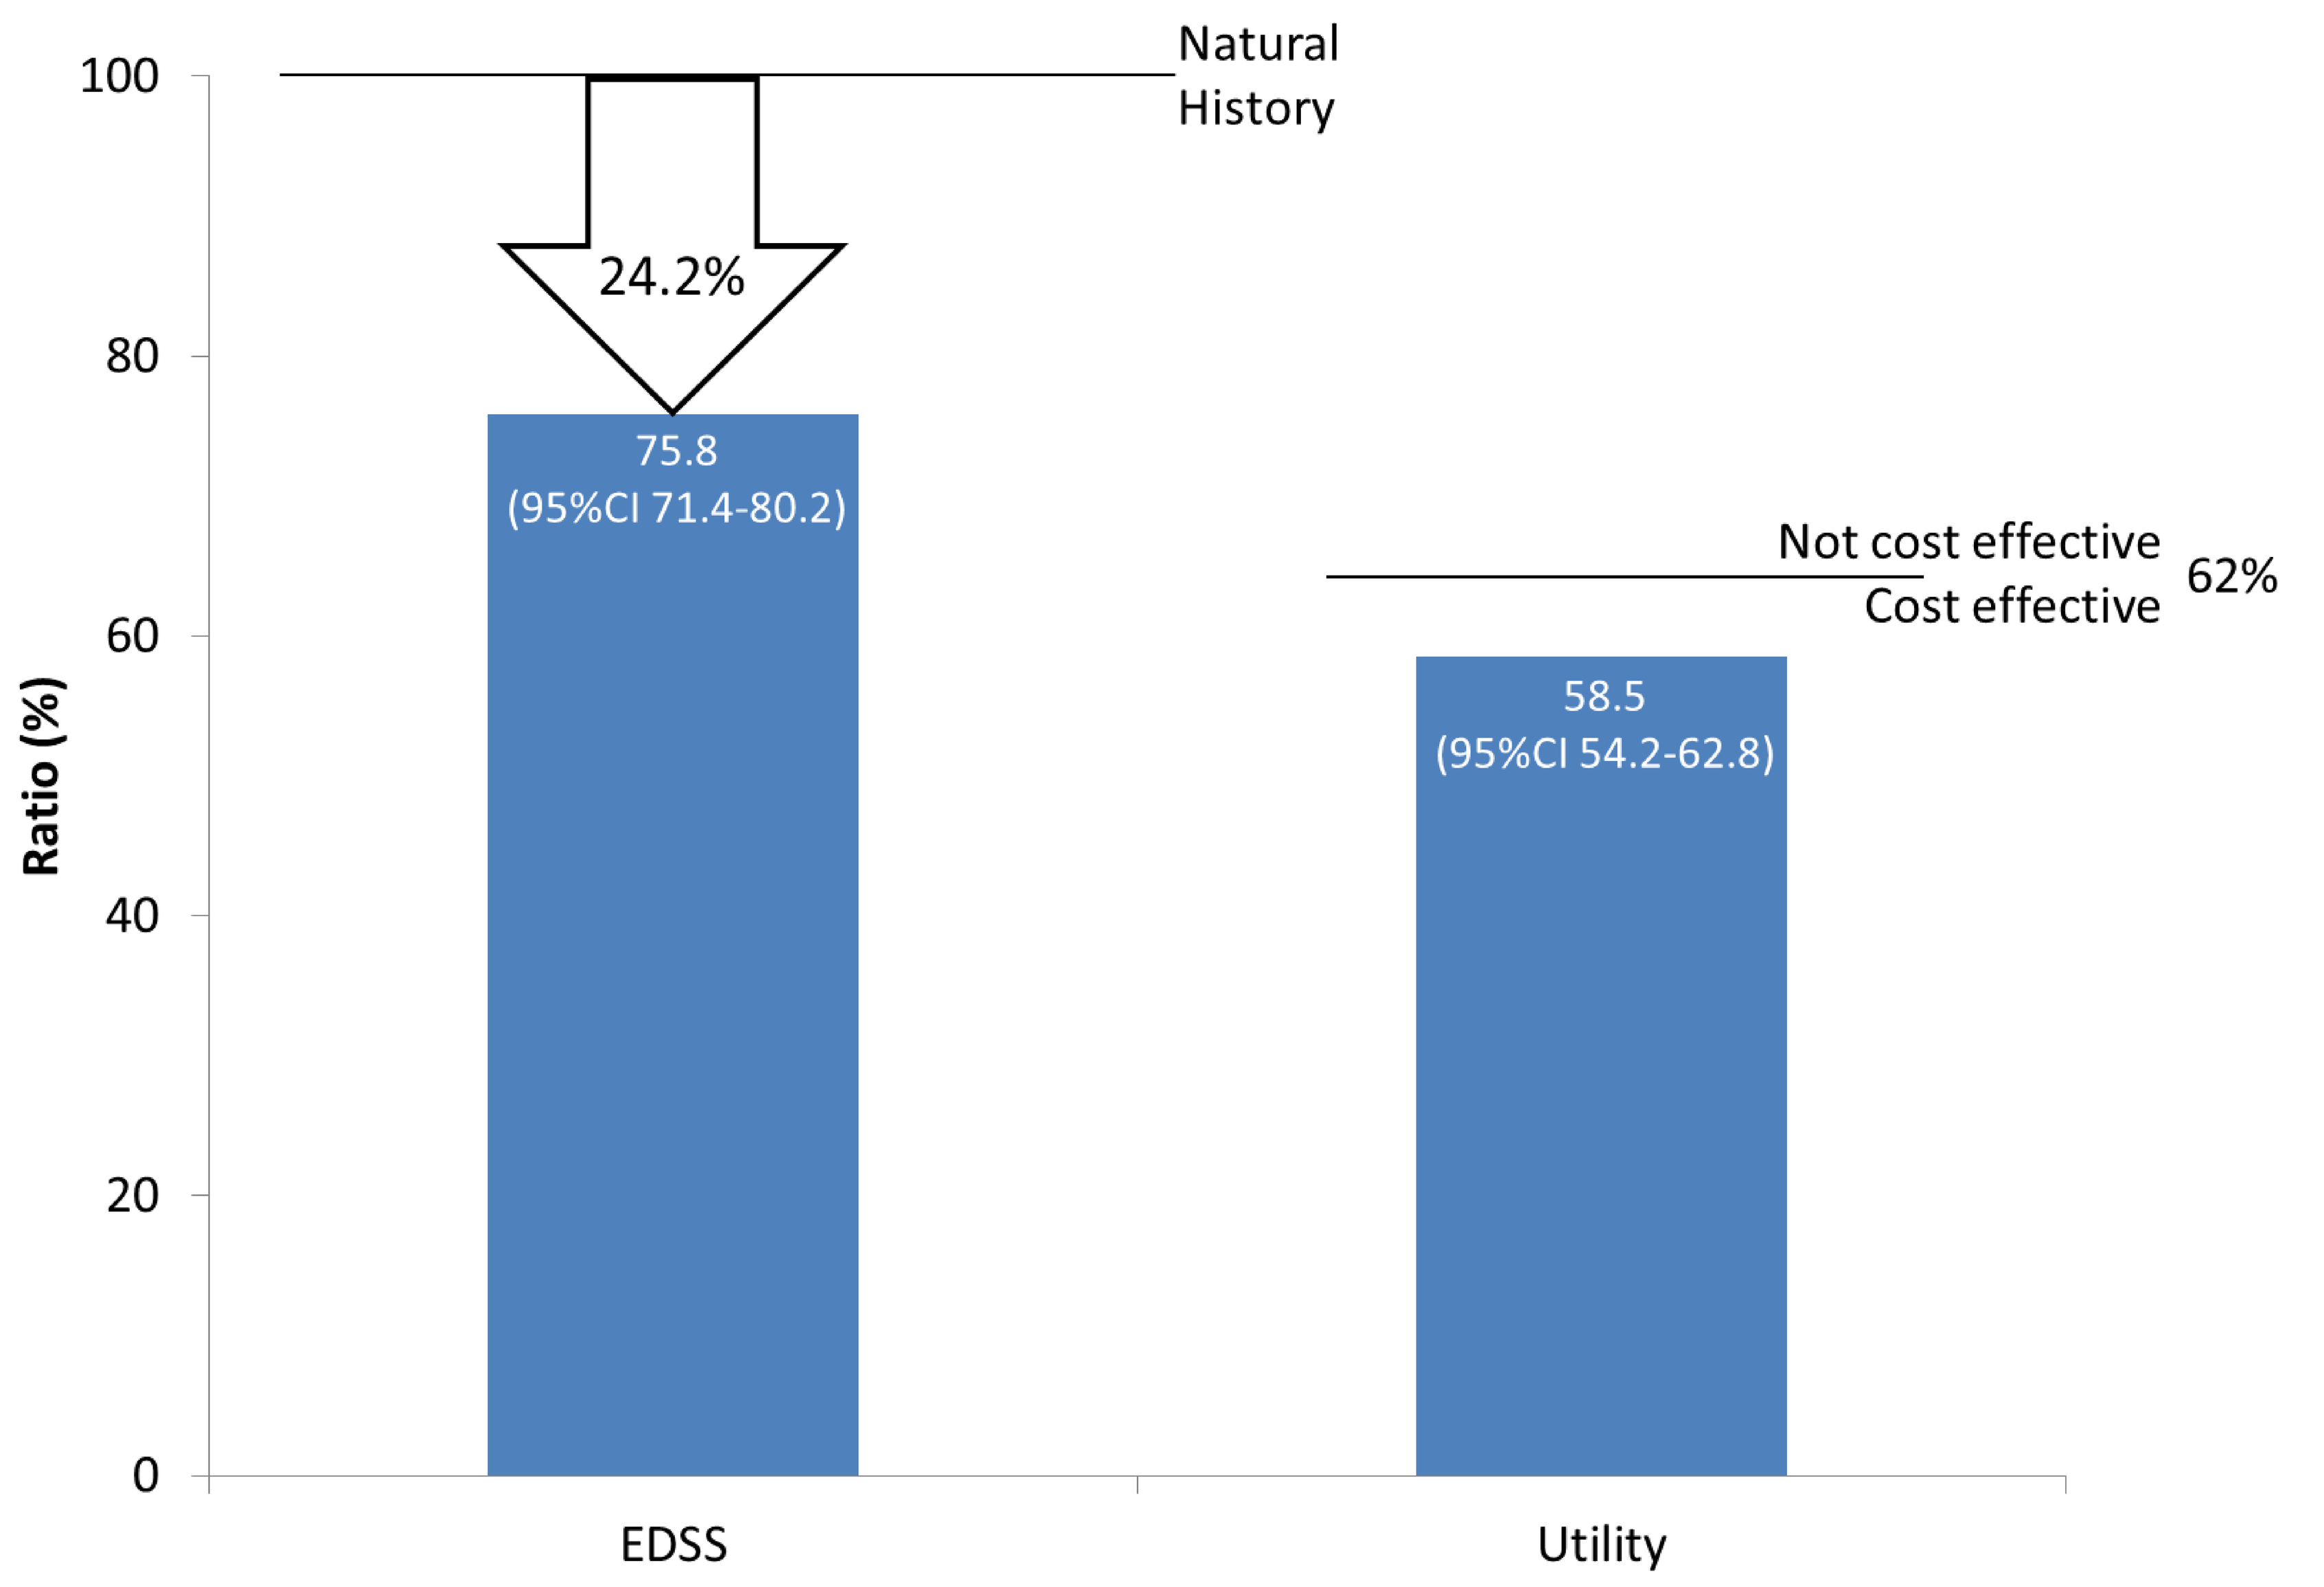
100
80
60
40
20
0
Ratio (%)
Natural
History
24.2%
75.8
(95%CI 71.4-80.2)
Not cost effective
Cost effective
62%
58.5
(95%CI 54.2-62.8)
EDSS
Utility

A ratio of less than 100% for EDSS implies slower than expected progression on DMT treatment compared to the untreated cohort. A utility progression ratio of 62% or lower means that utility progression for the drugs in aggregate is in line with, or slower than, the target cost effectiveness

OBJECTIVE

- To assess the clinical- and cost-effectiveness of GA, in terms of disability progression, using 6-year data from the RSS

METHODS

- A continuous Markov model with a 20-year time horizon was used to compare changes in EDSS and utility between patients on GA and the untreated comparator group
 - Patients receiving GA had fulfilled the Association of British Neurologists (ABN) 2001 criteria for DMT treatment, with treatment choice representing patient and physician preference at the time of prescribing (patients were not randomly assigned)
 - The comparator cohort used patient data from the BCMS database who met the same eligibility criteria (data collection 1980-1996)
- Primary outcomes were HRs (treated *vs* untreated) for accumulation of disability measured both as EDSS progression and as loss of utility
 - A ratio of less than 100% for EDSS implied slower than expected progression on treatment with GA compared to the untreated cohort
- Cost effectiveness of GA was expressed in two different ways:
 - The HR for EDSS progression was compared to a target HR derived from the RCTs of GA, with a pre-specified deviation of 10% about this target set, above or below which the price of GA would decrease or increase, respectively
 - The HR for EDSS progression was input into the cost-effectiveness calculator part of the model to determine whether the cost per QALY, based on a cost of GA *per* annum of £5,823 set at the inception of the scheme,[1] was above or below the £36,000 target

RESULTS

- 978 patients starting GA and 898 comparator patients were included in the analyses (Table 1)
- The cohorts had broadly similar baseline characteristics, although the GA cohort was older at disease onset (median 30 *vs* 28 years, respectively) and had a higher EDSS at baseline (median 3 *vs* 2) than the BCMS cohort

Table 1: Baseline characteristics of GA patients in RSS cohort and BCMS database

	GA cohort (n=978)	BCMS (n=898)
Sex		
- Men	223 (23%)	232 (26%)
- Women	755 (77%)	666 (74%)
Age at onset (years)		
- Mean	30.0 (8.05)	29.2 (8.7)
- Median	30 (24-35)	28 (23-35)
EDSS at baseline		
- Mean	3.07 (1.53)	2.44 (1.70)
- Median†	3 (2-4)	2 (1-3.5)
Mean follow-up	5.23 (1.33)	6.4 (3.5)

Data are n (%), mean (SD), or median (IQR). † EDSS scores are half-integers

- The model showed a 30.3% reduction in EDSS progression (HR 69.7) with GA therapy *vs* no treatment (Figure 2), which exceeded the target HR for cost effectiveness by over 10%
- GA therapy was also shown to reduce utility loss by 55.8% (HR 44.2) *vs* no treatment
- In absolute terms, patients who received GA had a mean EDSS score that was 0·404 units less than would have occurred without therapy (as predicted by the model)
- The resultant cost per QALY for GA was well below the target of £36,000

Figure 2: Progression and utility ratios for GA using 6-year RSS data and continuous Markov model

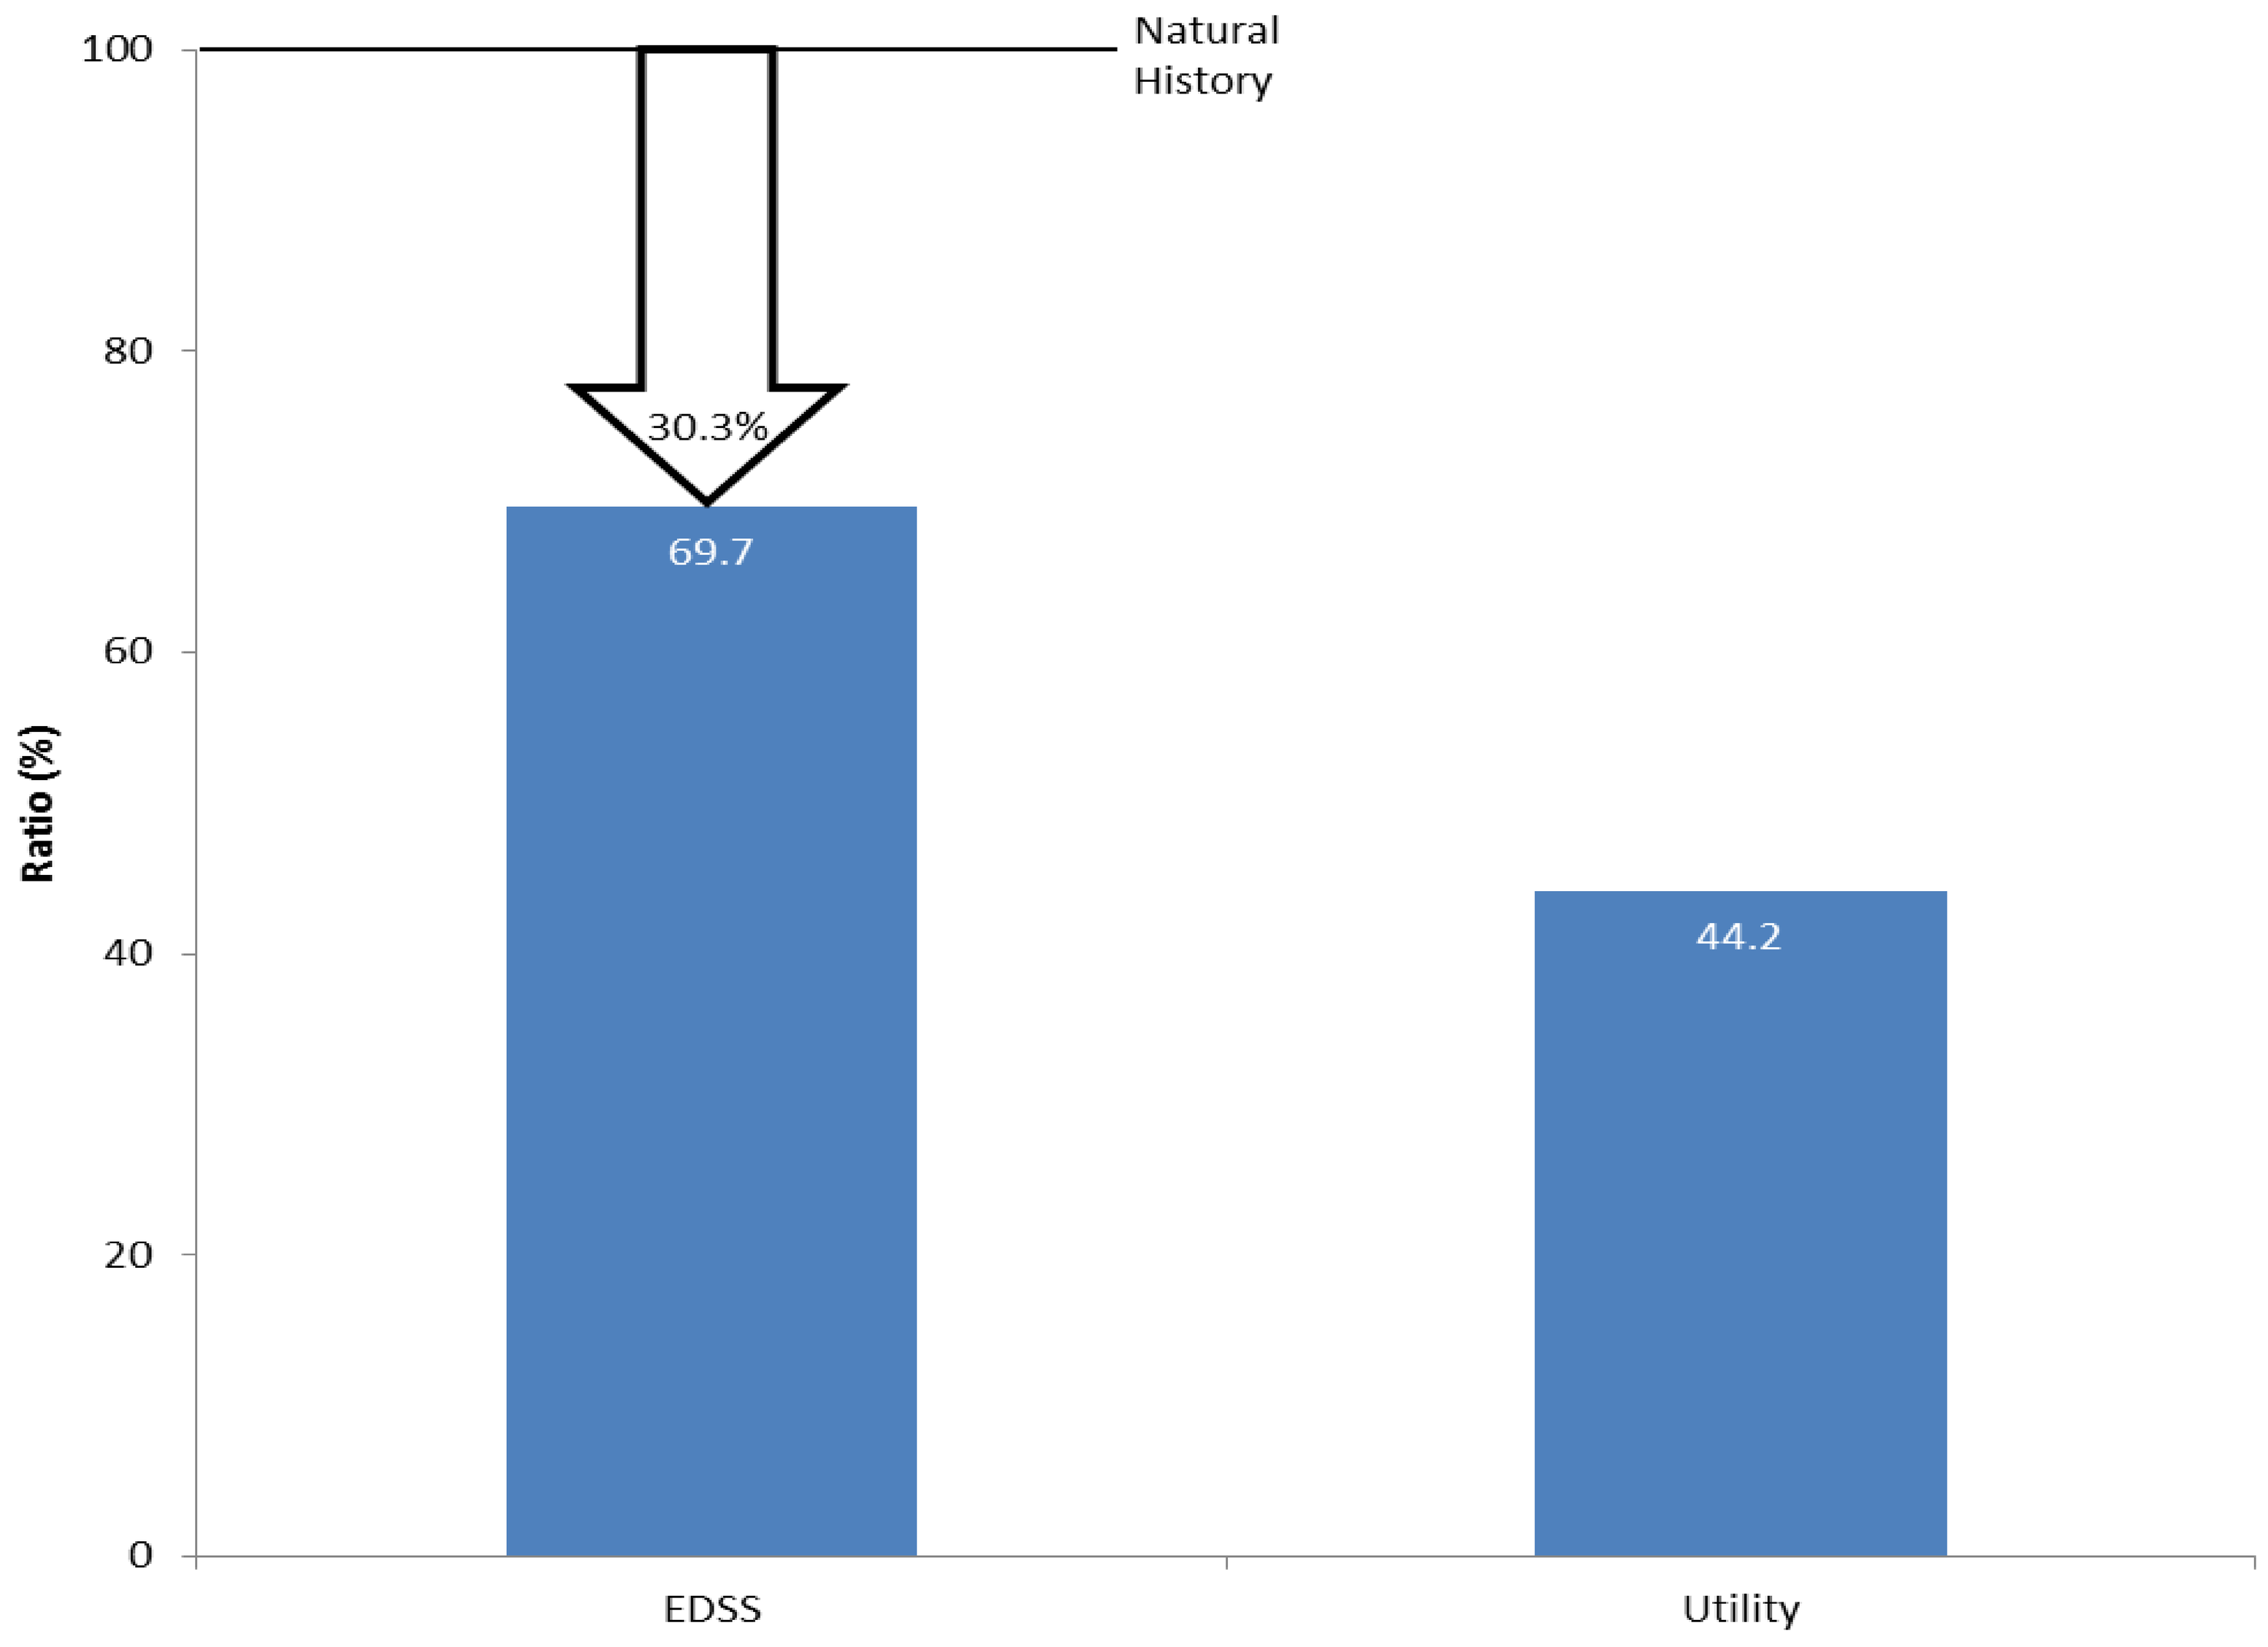
100
80
60
40
20
0
Ratio (%)
Natural
History
30.3%
69.7
44.2
EDSS
Utility

CONCLUSIONS

- This 6-year analysis supports the long-term efficacy of GA in terms of disability in patients with relapsing MS
- GA was shown to be more cost effective than predicted at the outset of the RSS, when modelled using the 6-year data over a 20-year time horizon
- Further insights into the long-term benefits of GA will come from the forthcoming analyses of the 8- and 10-year RSS data

REFERENCES

1. Department of Health. HSC 2002/004; Feb 2002.
2. Palace J *et al*. *Lancet Neurol* 2015; 14: 497-505.
3. MIMS, October 2015.

ACKNOWLEDGEMENTS

- The authors would like to thank Dr Jacqueline Palace (Oxford University Hospitals Trust, Oxford, UK) and Dr Martin Duddy (Newcastle upon Tyne Hospitals, Newcastle upon Tyne, UK) for reviewing the abstract prior to submission

DISCLOSURES

- Source of funding: Health Departments of England, Wales, Scotland and Northern Ireland, Teva Pharmaceuticals Industries, Biogen Idec, Merck Serono, Bayer Schering Pharmaceuticals, UK National Institute of Health Research's Health Technology Assessment Programme
- **Montu Sumra and Ewan Walters** are employees of Teva UK Limited. **John Fullarton** works for Strategen Limited who receive fees from Teva UK Limited to analyse the RSS data for GA

UK
Teva UK Limited

Teva UK Limited
Ridings Point, Whistler Drive, Castleford, WF10 5HX
T: 01977 628500 F: 01977 628799
www.tevauk.com

UK/CNS/15/0012
Date of Preparation: November 2015